61st ASH Annual Meeting & Exposition, December 7–10, 2019, Orlando, Florida, USA

Abstract 3745

Exposure-response relationship of the SMART-Ig Anti-hC5 antibody crovalimab (SKY59): Results from the umbrella phase 1/2 COMPOSER trial in healthy volunteers and patients with PNH

Alexandre Sostelly,[1] Simon Buatois,[1] Antoine Soubret,[1] Erica Winter,[2] Barbara Klughammer,[1] Joy C Hsu,[2] Gregor Jordan,[3] Christoph Bucher,[1] Jean Eric Charoin,[1] Keisuke Gotanda,[4] Kenji Shinomiya,[4] Zsolt Nagy,[5] Jens Panse,[6] Sung-Soo Yoon,[7] Régis Peffault de Latour,[8] Junichi Nishimura,[9] Alexander Röth[10]

[1]Roche Pharma Research and Early Development, Roche Innovation Center Basel, Basel, Switzerland; [2]Roche Pharma Research and Early Development, Roche Innovation Center New York, New York, NY; [3]Roche Pharma Research and Early Development, Roche Innovation Center Munich, Munich, Germany; [4]Chugai Pharmaceutical Co, Ltd, Tokyo, Japan; [5]Semmelweis Egyetem I.sz, Belgyógyászati Klinika, Budapest, Hungary; [6]University Hospital RWTH Aachen, Aachen, Germany; [7]Seoul National University Hospital, Seoul, South Korea; [8]CHU Paris-GH St-Louis Lariboisière F-Widal Hospital, Paris, France; [9]Osaka University Hospital, Osaka, Japan; [10] Department of Hematology, University Hospital Essen, Essen, Germany

PNH AND CROVALIMAB

Disease background and current treatment limitations

- Paroxysmal nocturnal hemoglobinuria (PNH) is a clonal, nonmalignant, hematologic disorder.[1]
 - Mutation in the phosphatidylinositol glycan anchor biosynthesis class A (*PIGA*) gene affects the complement pathway, making erythrocytes susceptible to complement-mediated hemolysis.[1]
- Current standard of care for PNH consists of inhibition of complement activity by blocking complement component 5 (C5) with either eculizumab[2] or ravulizumab.[3]
- Current treatment limitations include extravacular hemolysis, the need for large antibody doses, breakthrough hemolysis due to non-sustained C5 inhibition and the lack of efficacy in patients with C5 mutational variants[4,5]

Crovalimab: a novel, optimized anti-C5 antibody

- Crovalimab, a novel antihuman C5 antibody, has been engineered with Sequential Monoclonal Antibody Recycling Technology–Immunoglobulin (SMART-Ig) (**Figure 1**).[4,6-8]
- SMART allows for:
 - Reducing the drug amount required to achieve full complement inhibition through pH-dependent binding and target disposal.
 - Increasing half-life through antibody recycling and neonatal Fc receptor (FcRn) engineering.

Figure 1. Crovalimab properties and mechanism of action[7,9]

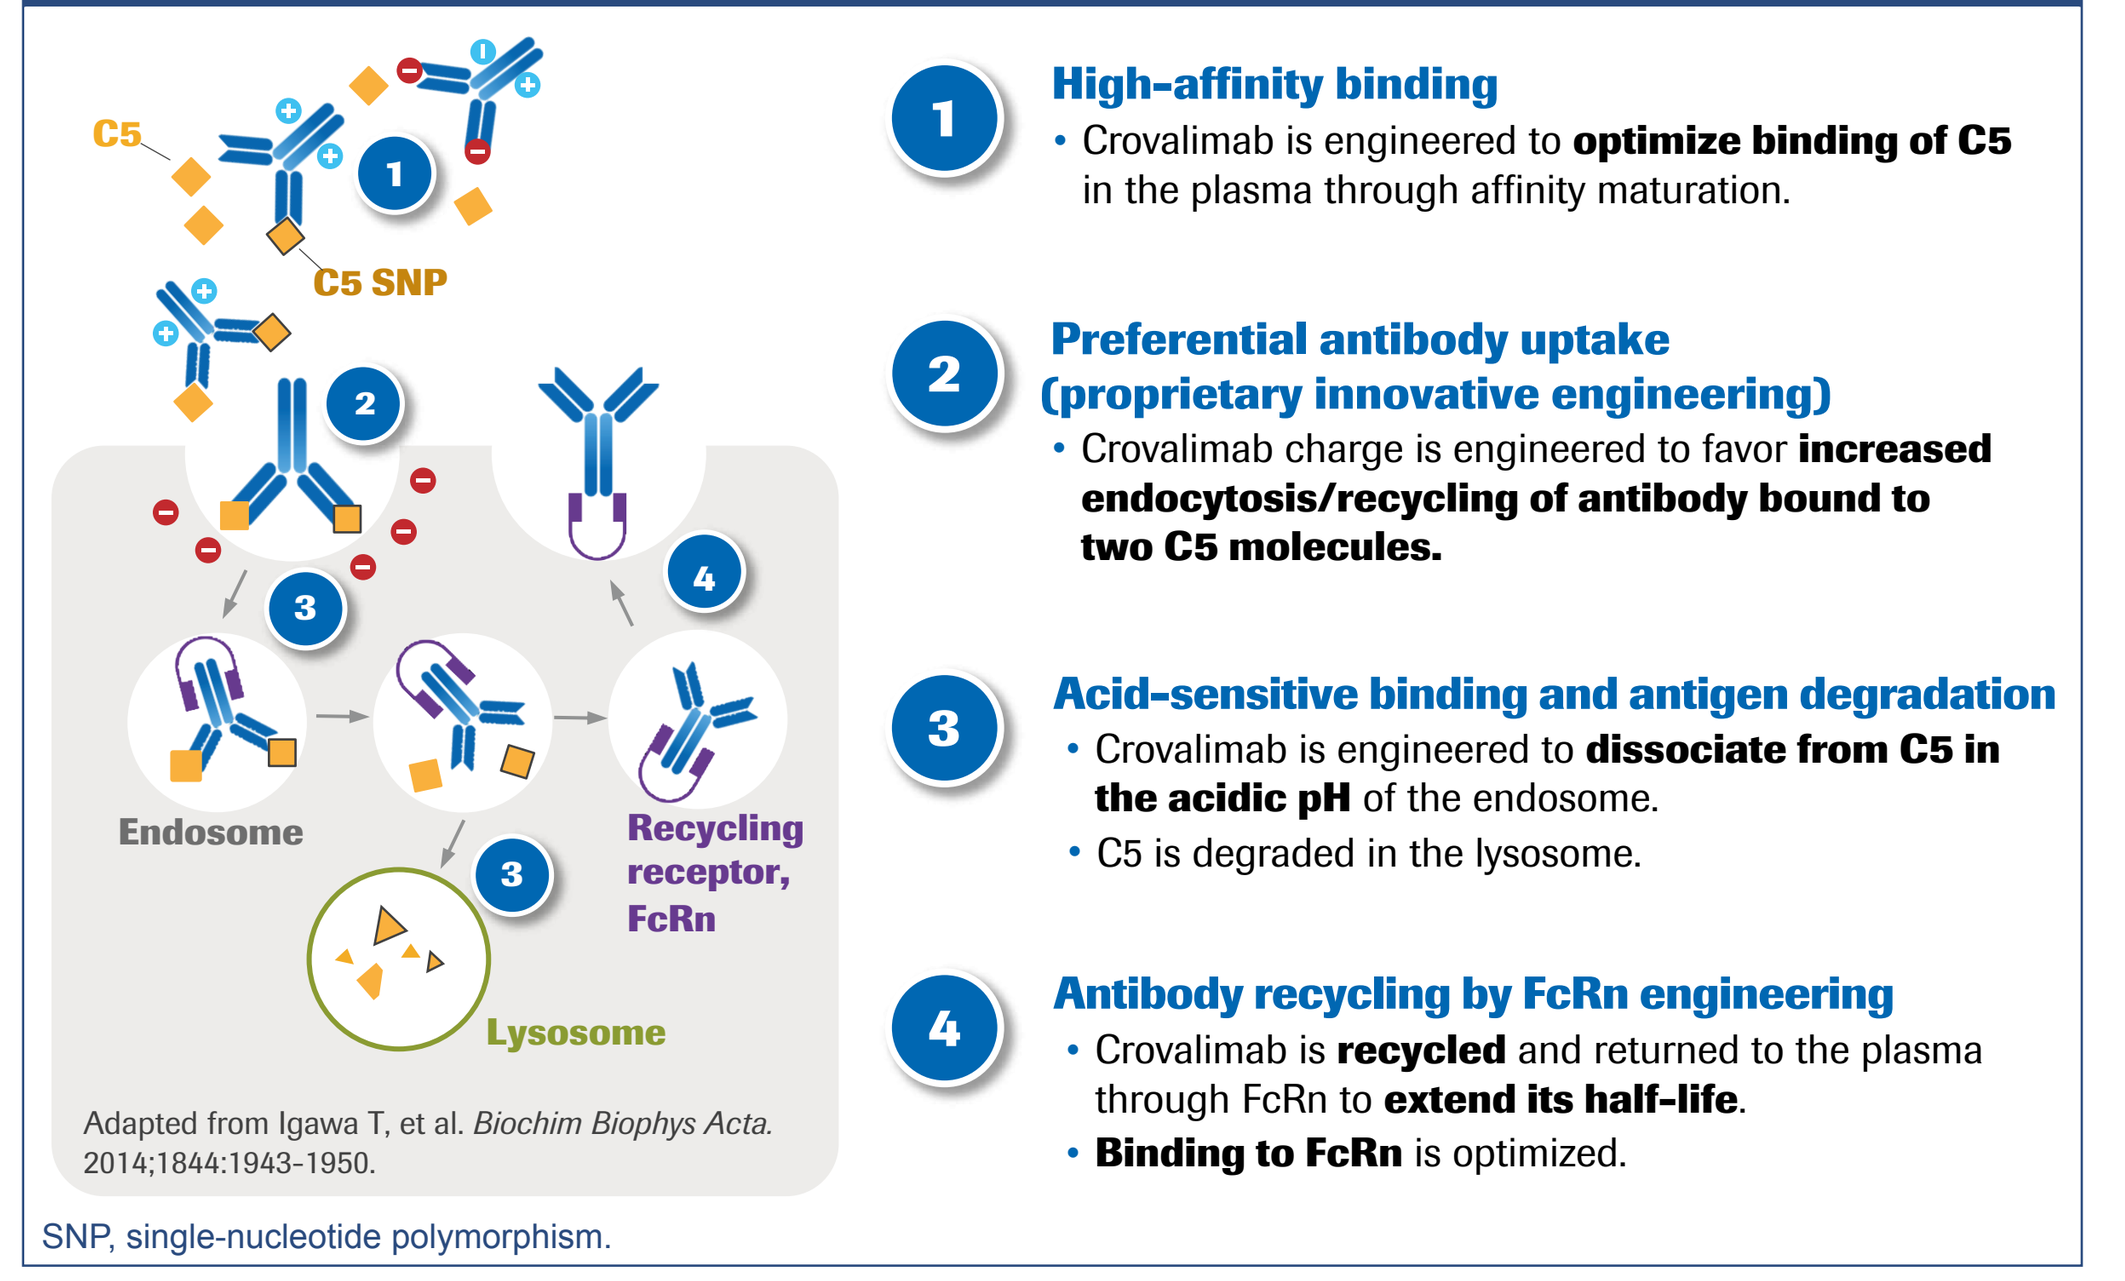
C5
C5 SNP
Endosome
Recycling receptor, FcRn
Lysosome

Adapted from Igawa T, et al. *Biochim Biophys Acta*. 2014;1844:1943-1950.

1. **High-affinity binding**
 - Crovalimab is engineered to **optimize binding of C5** in the plasma through affinity maturation.
2. **Preferential antibody uptake (proprietary innovative engineering)**
 - Crovalimab charge is engineered to favor **increased endocytosis/recycling of antibody bound to two C5 molecules.**
3. **Acid-sensitive binding and antigen degradation**
 - Crovalimab is engineered to **dissociate from C5 in the acidic pH** of the endosome.
 - C5 is degraded in the lysosome.
4. **Antibody recycling by FcRn engineering**
 - Crovalimab is **recycled** and returned to the plasma through FcRn to **extend its half-life**.
 - **Binding to FcRn** is optimized.

SNP, single-nucleotide polymorphism.

COMPOSER STUDY

- COMPOSER (NCT03157635) is a phase 1/2, four-part adaptive clinical trial to establish the pharmacokinetics (PK), pharmacodynamics (PD), and optimal dose of crovalimab that would completely inhibit C5 in patients with PNH (**Figure 2**).

Figure 2. COMPOSER study design[8]

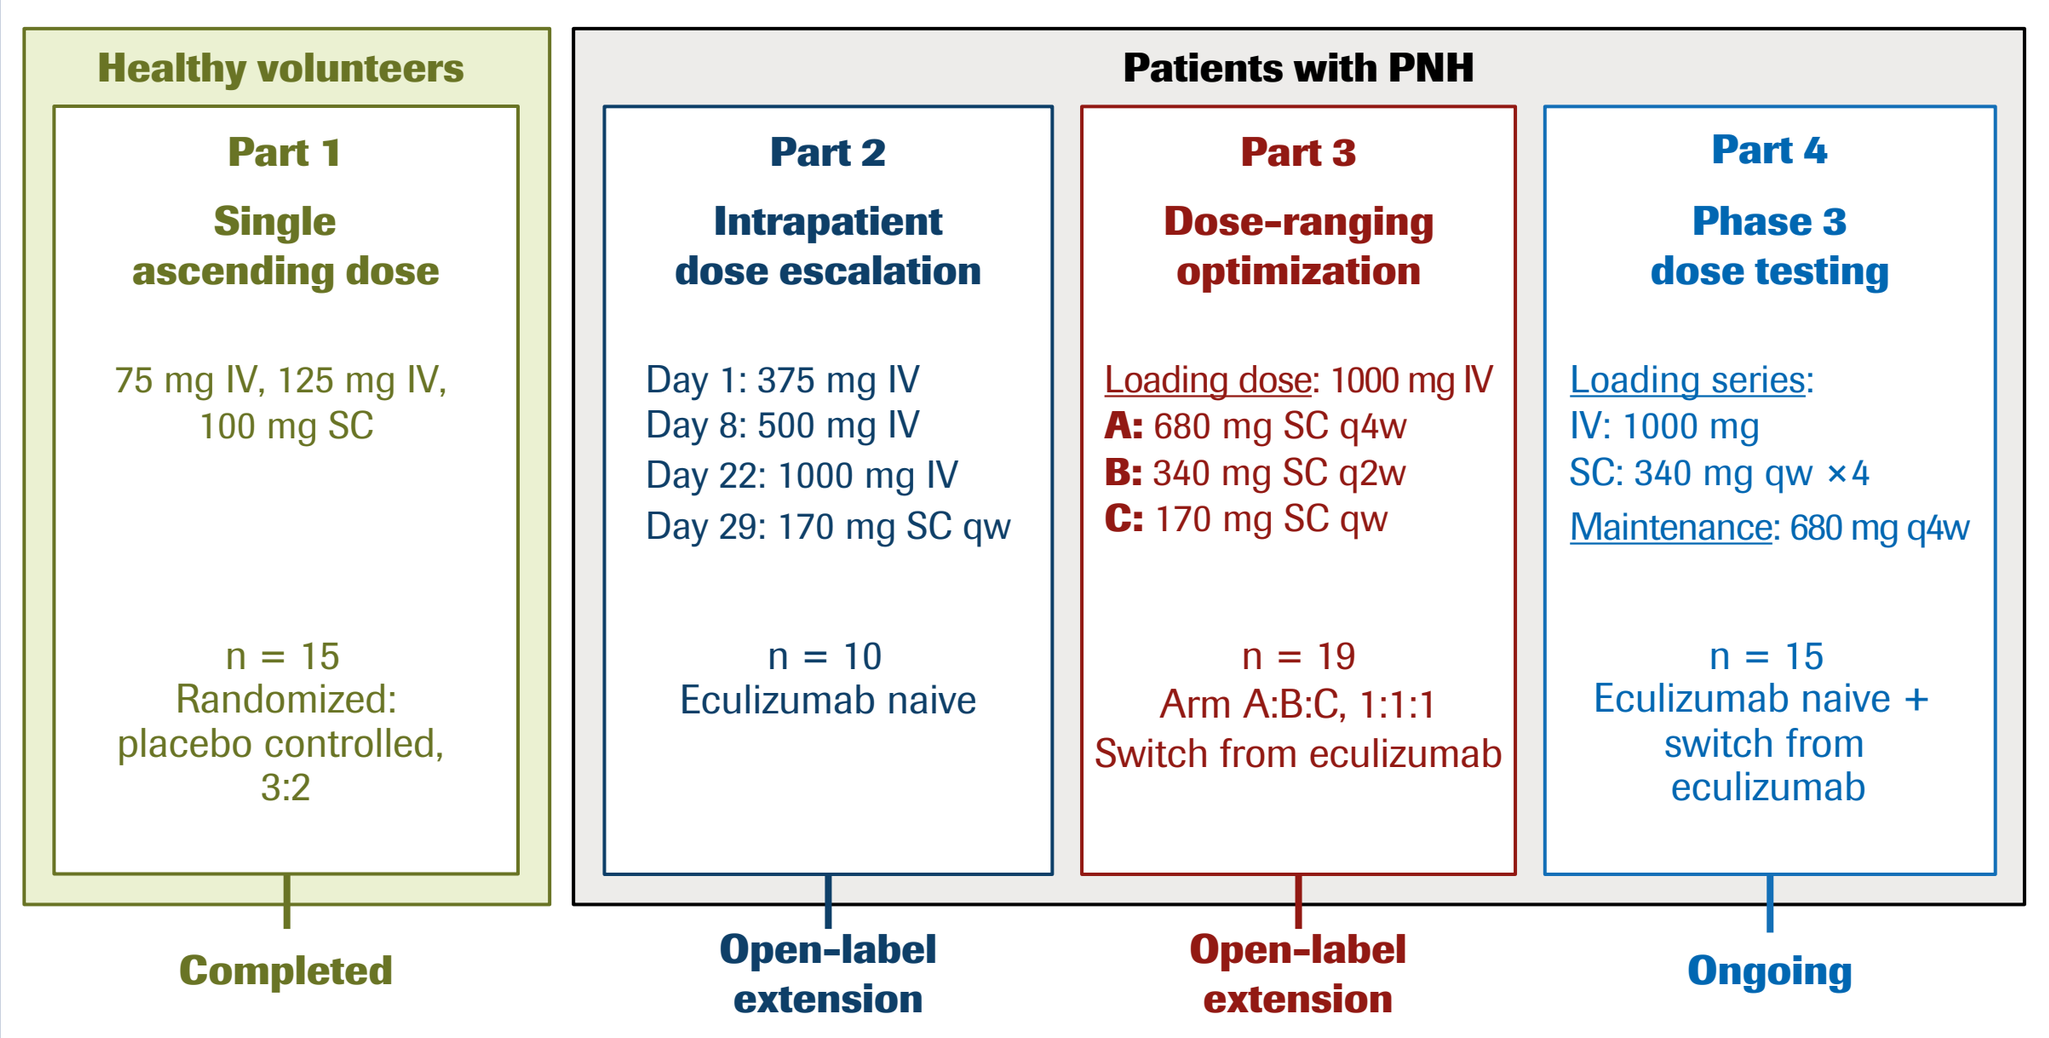
Healthy volunteers
Part 1
Single ascending dose
75 mg IV, 125 mg IV, 100 mg SC
n = 15
Randomized: placebo controlled, 3:2
Completed
Patients with PNH
Part 2
Intrapatient dose escalation
Day 1: 375 mg IV
Day 8: 500 mg IV
Day 22: 1000 mg IV
Day 29: 170 mg SC qw
n = 10
Eculizumab naive
Open-label extension
Part 3
Dose-ranging optimization
Loading dose: 1000 mg IV
A: 680 mg SC q4w
B: 340 mg SC q2w
C: 170 mg SC qw
n = 19
Arm A:B:C, 1:1:1
Switch from eculizumab
Open-label extension
Part 4
Phase 3 dose testing
Loading series:
IV: 1000 mg
SC: 340 mg qw ×4
Maintenance: 680 mg q4w
n = 15
Eculizumab naive + switch from eculizumab
Ongoing

IV, intravenous; SC, subcutaneous; qw, once a week; q2w, every 2 weeks; q4w, every 4 weeks.

- Crovalimab concentrations were measured using a validated enzyme-linked immunosorbent assay.
- Free C5 concentrations were measured using a fluorescent-based, ligand-binding assay.
- A population PK model was developed using all available data to describe the crovalimab concentration-time profiles.
- Crovalimab PD was assessed by evaluating the extent and duration of terminal complement inhibition, quantified using a validated, functional, ex vivo, liposome immunoassay (LIA).
- Relationships between crovalimab PK and PD were analyzed using graphic analysis.

CROVALIMAB PK/PD IN PATIENTS WITH PNH

- Crovalimab exposure over time in patients enrolled in parts 2 and 3 of COMPOSER is shown in **Figure 3**.
- Because eculizumab and crovalimab bind different C5 epitopes, switching from eculizumab to crovalimab induces formation of drug-target-drug complexes (DTDCs) consisting of crovalimab, C5, and eculizumab.
- In patients enrolled in part 3 who were pretreated with eculizumab, a transient decrease in crovalimab exposure occurred due to the formation of DTDCs upon switching to crovalimab.
- DTDCs were observed in all part 3 patients for approximately 10 weeks.
- The LIA assay showed rapid achievement of full complement activity inhibition when patients were dosed with crovalimab (**Figure 4**).
 - In all patients with PNH, complete complement inhibition (defined as LIA < 10 U/mL, the lower limit of quantitation of the assay) was achieved immediately after the IV loading dose and was maintained across the different dosing intervals in the majority of patients (**Figure 4**).
- Complete inhibition of free C5 (defined as free C5 < 0.05 μg/mL) was also achieved after crovalimab infusion and was maintained over time for all dosing intervals (**Figure 5**).

Figure 3. Mean crovalimab exposure in patients with PNH

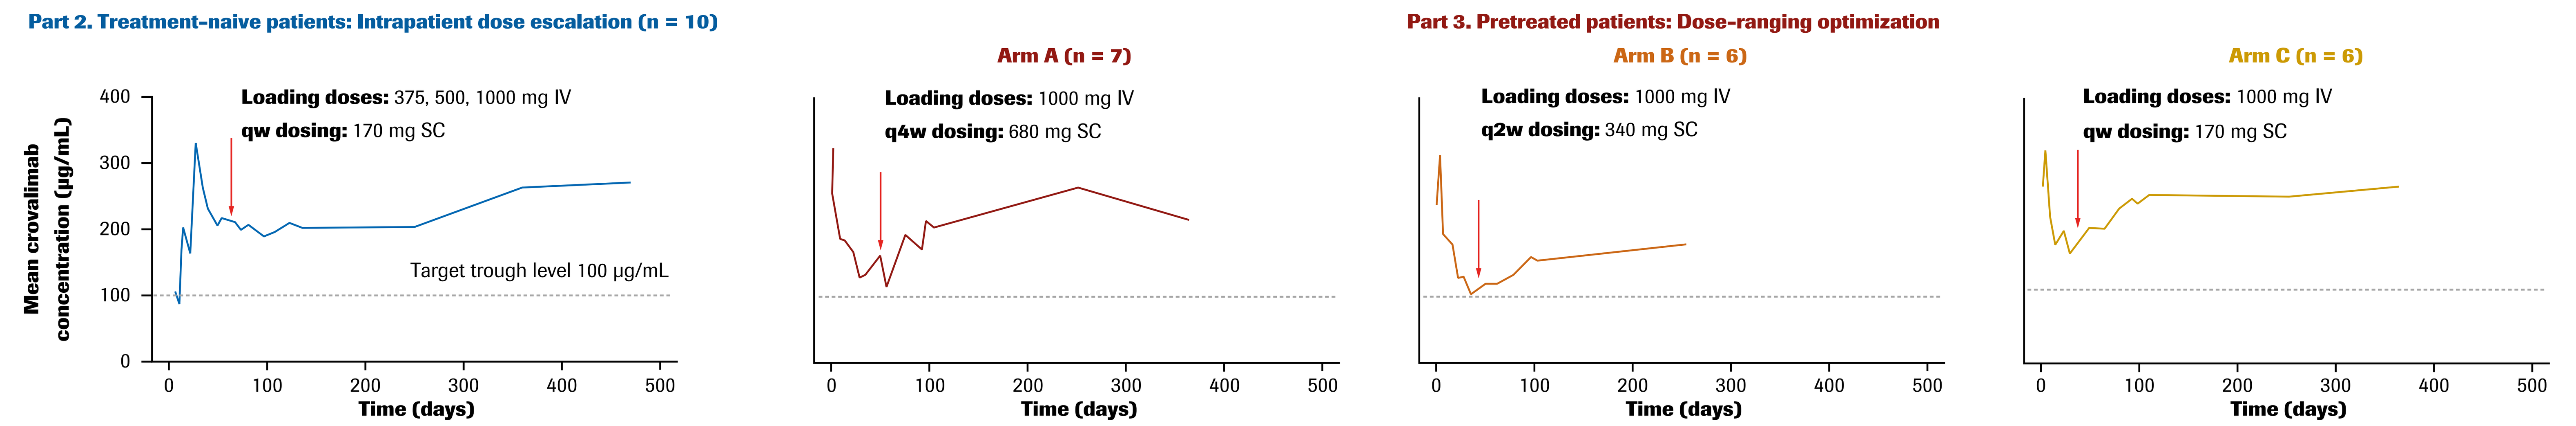
Part 2. Treatment-naive patients: Intrapatient dose escalation (n = 10)
Part 3. Pretreated patients: Dose-ranging optimization
Arm A (n = 7)
Arm B (n = 6)
Arm C (n = 6)
Loading doses: 375, 500, 1000 mg IV
qw dosing: 170 mg SC
Loading doses: 1000 mg IV
q4w dosing: 680 mg SC
Loading doses: 1000 mg IV
q2w dosing: 340 mg SC
Loading doses: 1000 mg IV
qw dosing: 170 mg SC
Target trough level 100 μg/mL
Mean crovalimab concentration (μg/mL)
Time (days)

Red arrows indicate the start of qw, q4w, or q2w dosing.

Figure 4. LIA time course showing mean terminal complement activity

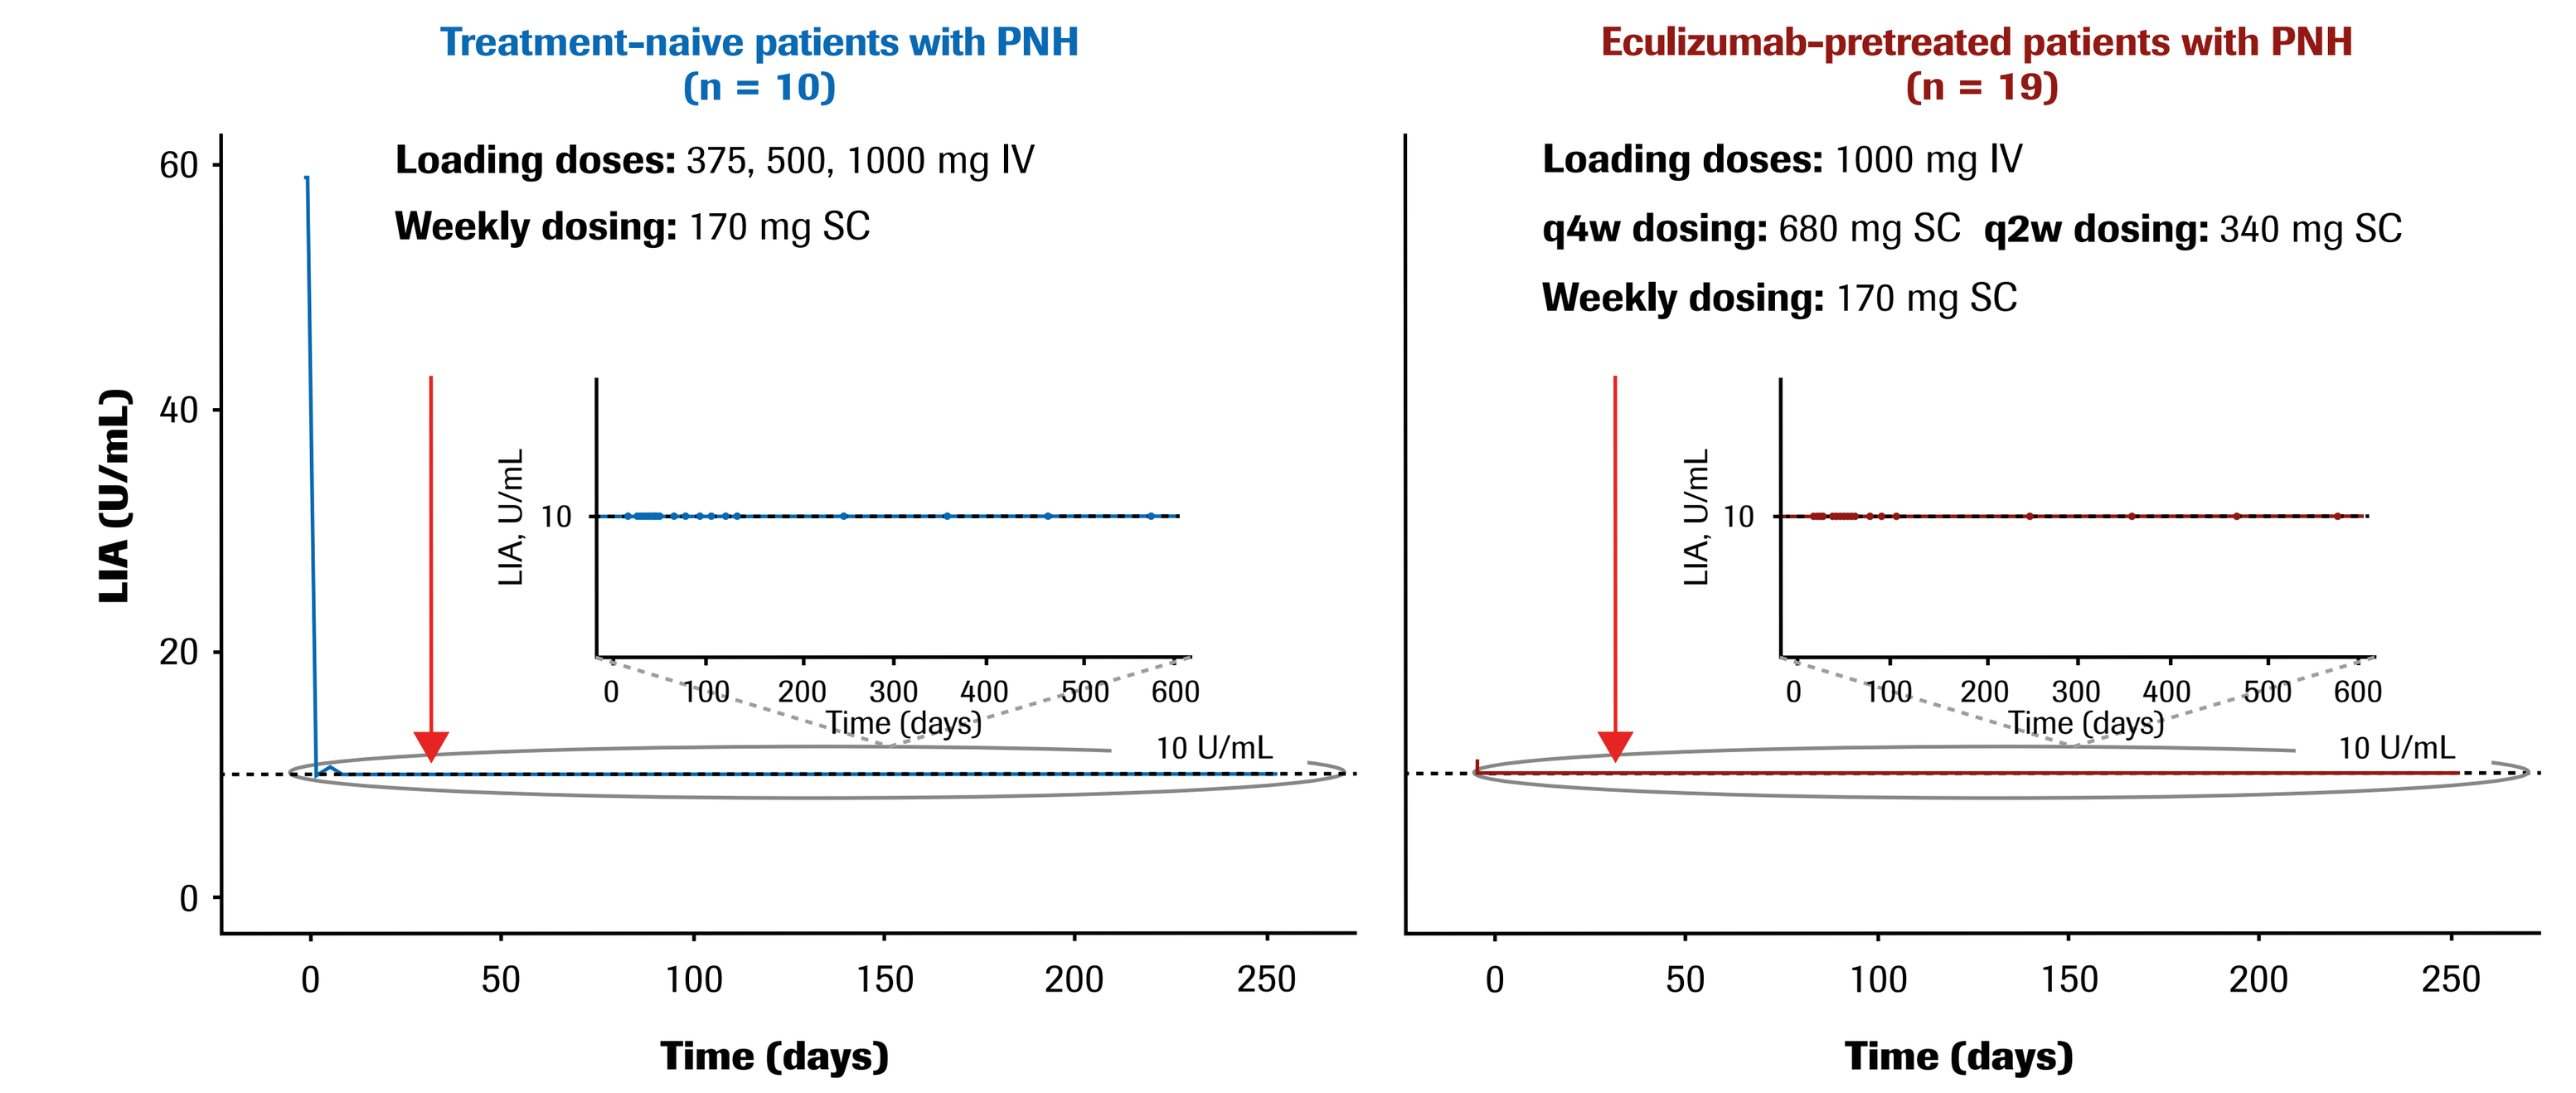
Treatment-naive patients with PNH (n = 10)
Loading doses: 375, 500, 1000 mg IV
Weekly dosing: 170 mg SC
Eculizumab-pretreated patients with PNH (n = 19)
Loading doses: 1000 mg IV
q4w dosing: 680 mg SC q2w dosing: 340 mg SC
Weekly dosing: 170 mg SC
LIA (U/mL)
LIA, U/mL
10 U/mL
Time (days)

Red arrows indicate the start of qw, q4w, or q2w dosing.

Figure 5. Time course of free C5 concentrations after initiation of crovalimab in patients with PNH who were (A) treatment naive and (B) eculizumab pre-treated

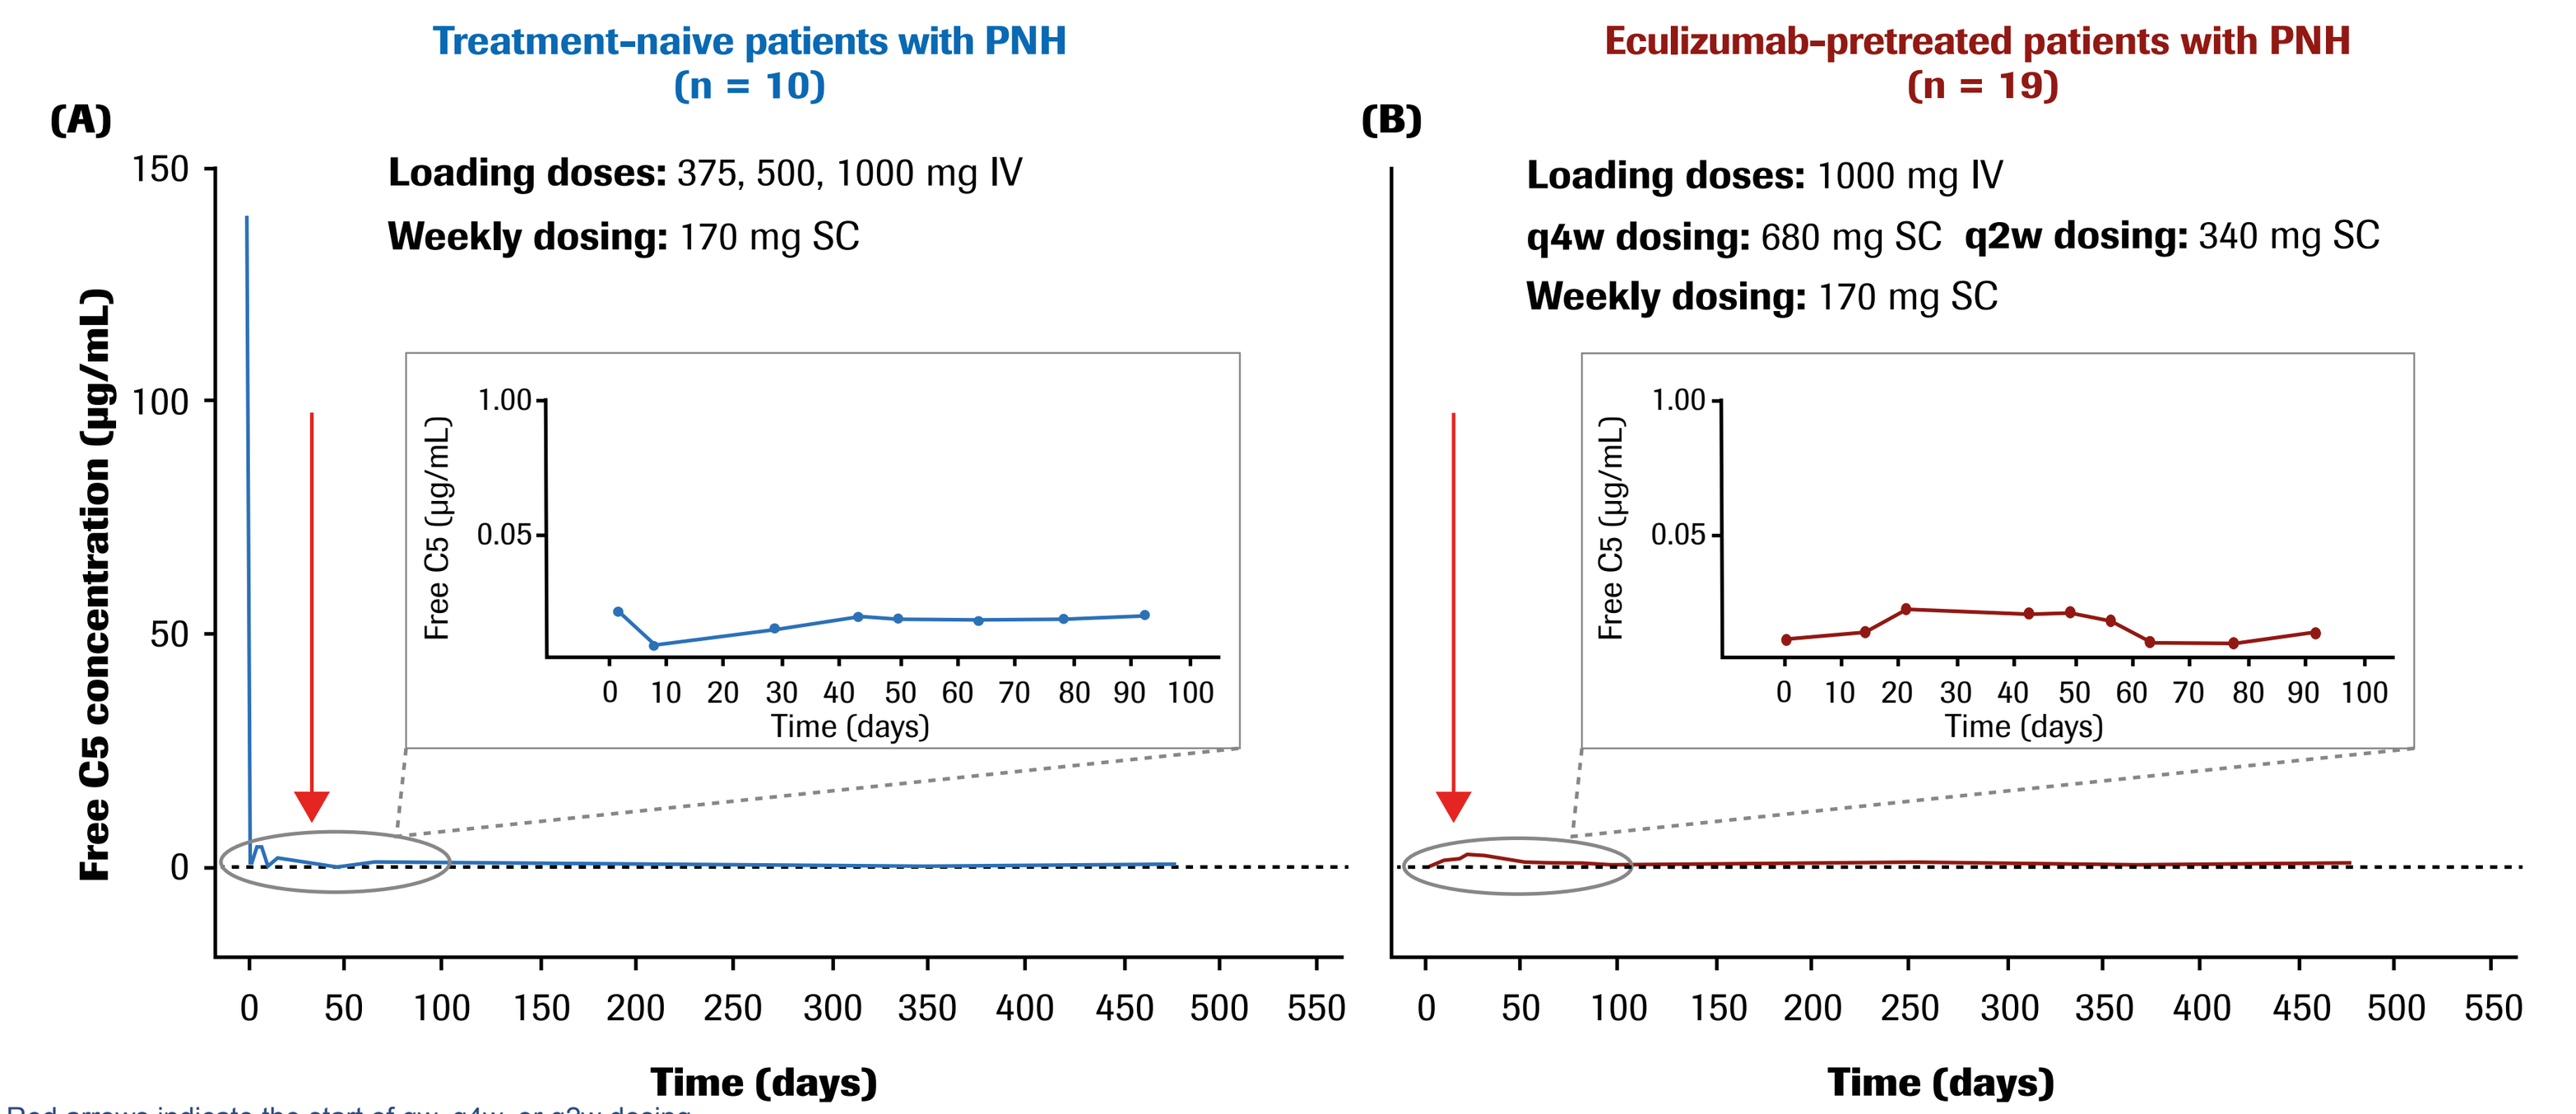
(A)
Treatment-naive patients with PNH (n = 10)
Loading doses: 375, 500, 1000 mg IV
Weekly dosing: 170 mg SC
(B)
Eculizumab-pretreated patients with PNH (n = 19)
Loading doses: 1000 mg IV
q4w dosing: 680 mg SC q2w dosing: 340 mg SC
Weekly dosing: 170 mg SC
Free C5 concentration (μg/mL)
Free C5 (μg/mL)
Time (days)

Red arrows indicate the start of qw, q4w, or q2w dosing.

POPULATION PK MODEL TO DESCRIBE CROVALIMAB CONCENTRATION-TIME PROFILE

- The PK of crovalimab was best described by a two-compartment disposition model with first-order elimination and absorption (**Figure 6**).
- To describe the PK in patients pretreated with eculizumab, elimination of crovalimab was modeled as a combination of the first-order elimination used for patients who were treatment naive and a faster clearance that decreases exponentially over time.
- Body weight was introduced using allometry on the clearances and volumes of the distribution.

Figure 6. Population PK model for crovalimab

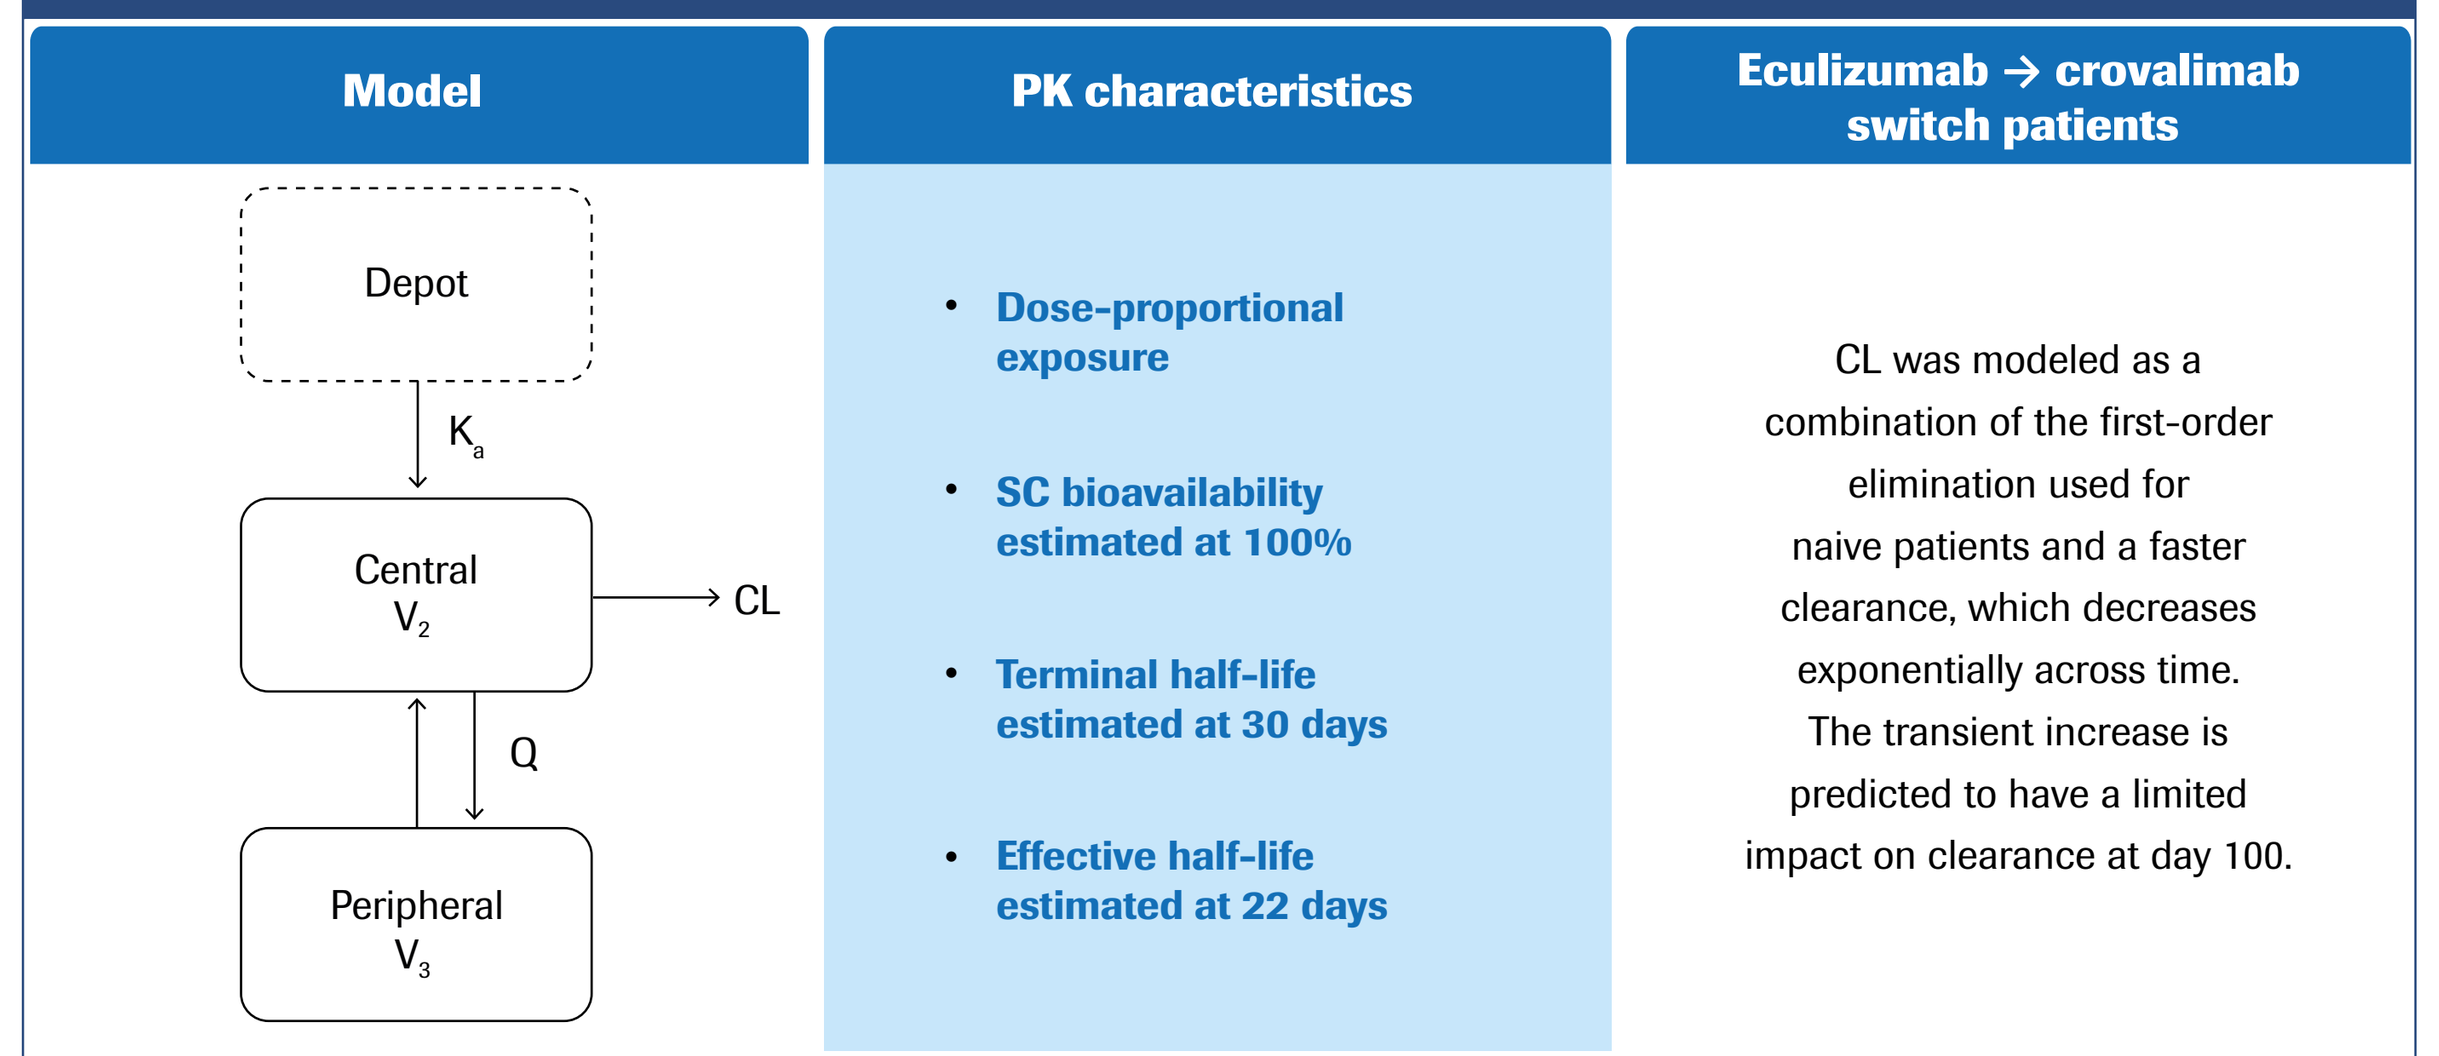
Model
Depot
K_a
Central V_2
CL
Q
Peripheral V_3
PK characteristics
Dose-proportional exposure
SC bioavailability estimated at 100%
Terminal half-life estimated at 30 days
Effective half-life estimated at 22 days
Eculizumab → crovalimab switch patients
CL was modeled as a combination of the first-order elimination used for naive patients and a faster clearance, which decreases exponentially across time. The transient increase is predicted to have a limited impact on clearance at day 100.

CL, total clearance from plasma; K_a, first-order absorption rate constant; Q, intercompartmental clearance; V, apparent volume.

RELATIONSHIP BETWEEN CROVALIMAB EXPOSURE, C5, AND COMPLEMENT ACTIVITY

- When the PK and PD data from parts 1 to 3 of COMPOSER were pooled, crovalimab was shown to induce a concentration-dependent inhibition of free C5 (**Figure 7A**) and of serum hemolytic activity (**Figure 7B**) that closely correlated with terminal complement inhibition.
- At the crovalimab concentration of approximately 100 μg/mL, complete terminal complement inhibition was achieved (**Figure 7B**).

Figure 7. Exposure-complement activity relationship of crovalimab in COMPOSER: relationship between total crovalimab concentrations and (A) free C5 or (B) LIA

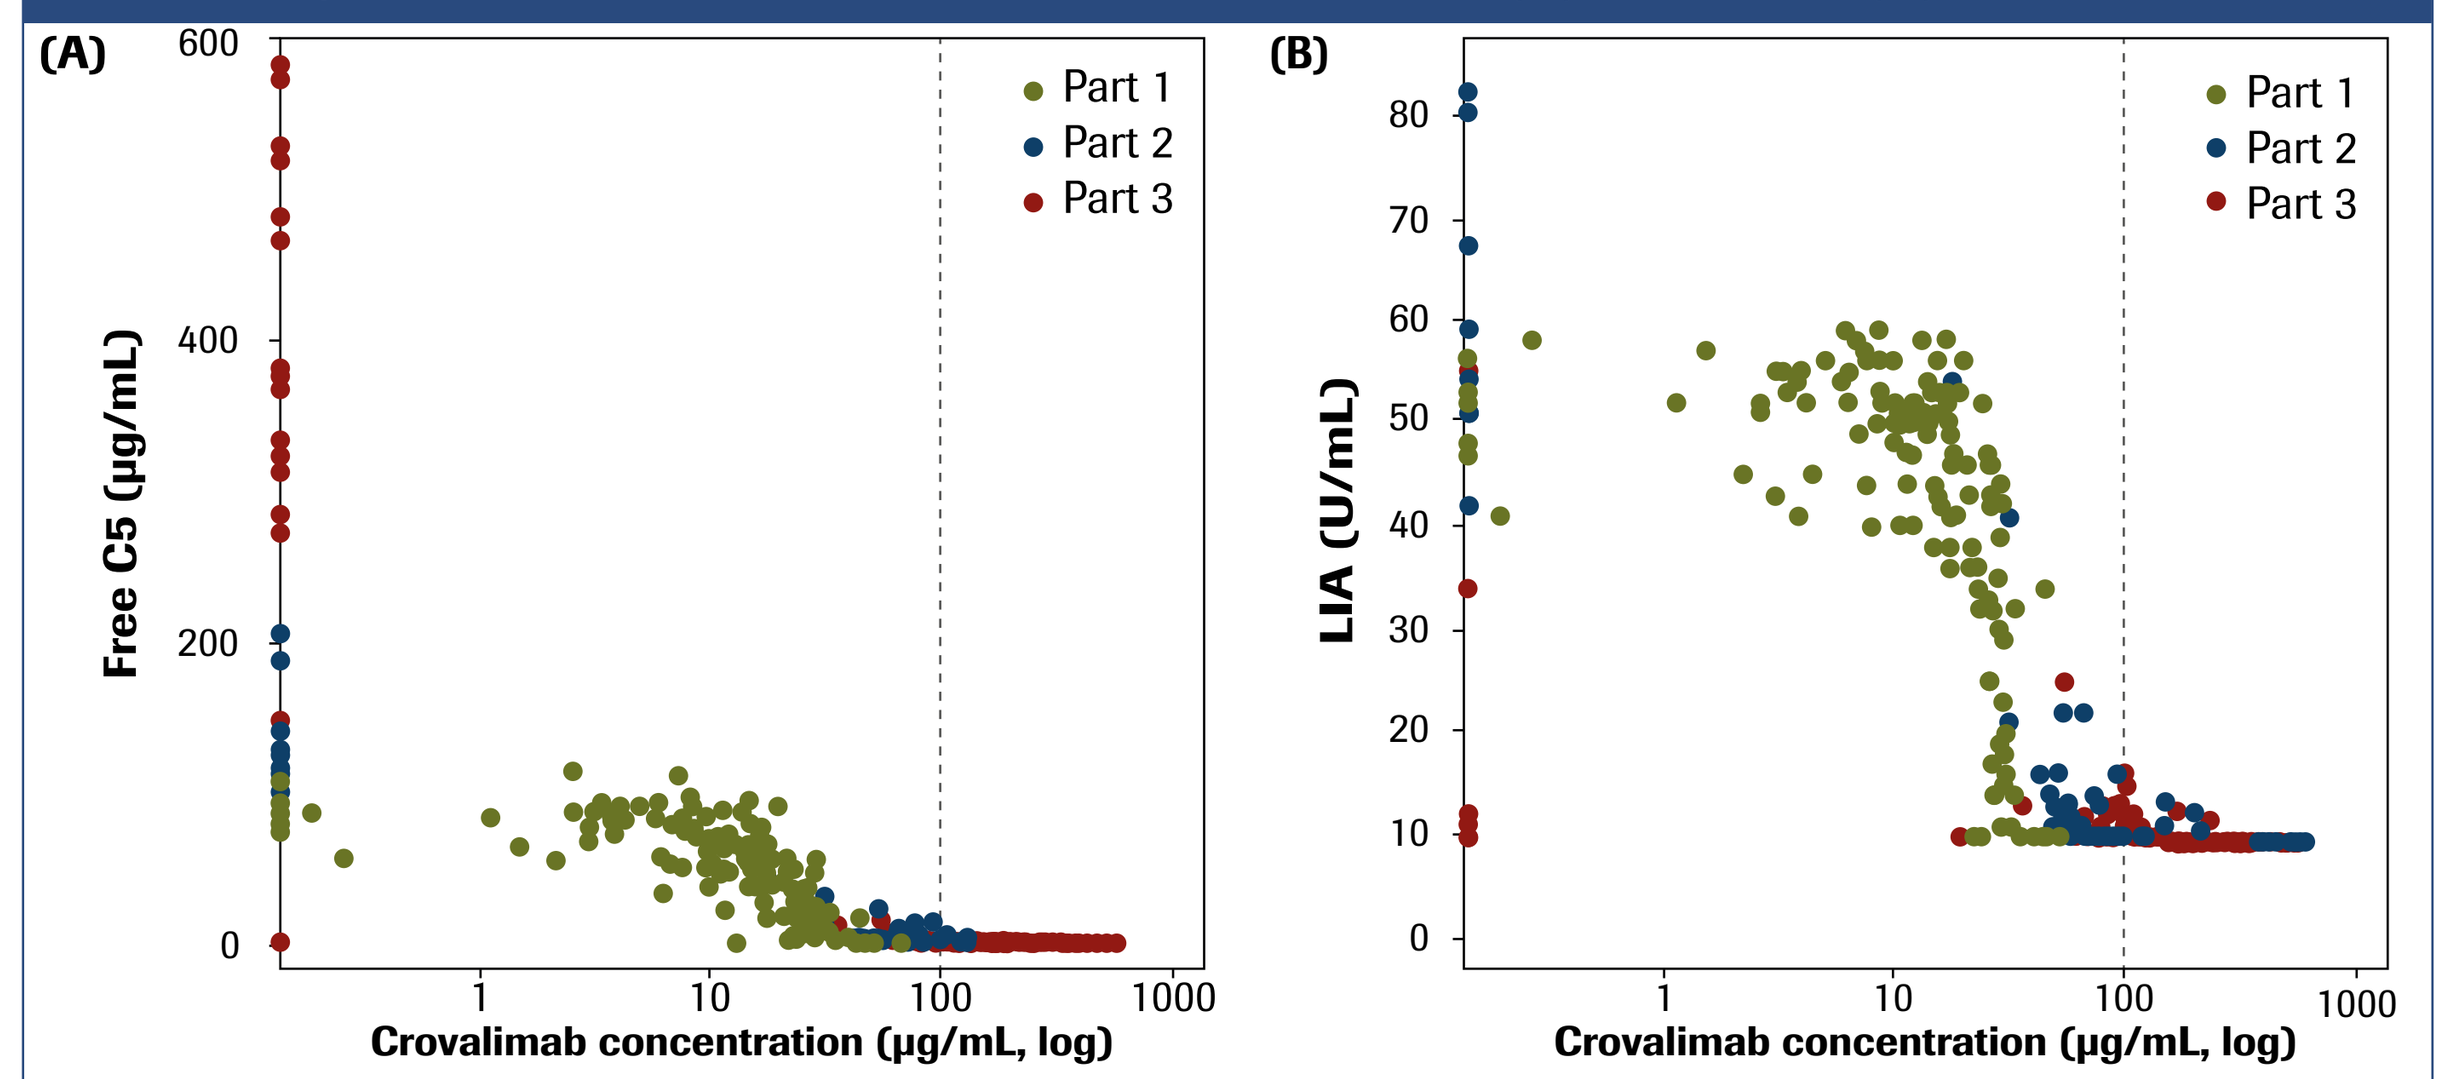
(A)
(B)
Free C5 (μg/mL)
LIA (U/mL)
Part 1
Part 2
Part 3
Crovalimab concentration (μg/mL, log)

Crovalimab concentration is shown on a logarithmic scale. The vertical line marks the PD threshold of 100 μg/mL crovalimab. Part 1 involved healthy volunteers; part 2, patients with PNH who were treatment naive; part 3, patients with PNH who switched from eculizumab.

USE OF SIMULATIONS TO OPTIMIZE CROVALIMAB DOSING

- Simulated concentration-time profiles of the crovalimab doses administered in part 3 (**Figure 8**) showed that the dosing regimen was potentially suboptimal in patients who switched from eculizumab to crovalimab, related to the transient formation of DTDCs.
- These patients were transiently at risk of having crovalimab fall below the target concentration identified from the exposure-response characterizations.
- Based on PK modeling, an optimized dosing regimen is being evaluated in part 4 of COMPOSER:
 - 1000 mg IV on day 1
 - 340 mg SC on days 2, 8, 15, and 22
 - 680 mg SC on day 29 and then q4w
- With this optimized regimen, even patients who switch from eculizumab are expected to achieve the target crovalimab concentration needed for complete terminal inhibition of complement (**Figure 9**).

Figure 8. Simulated concentration-time profiles of crovalimab based on doses used in Part 3 of COMPOSER in (A) patients with PNH who were treatment naive and (B) patients with PNH who switched from eculizumab treatment

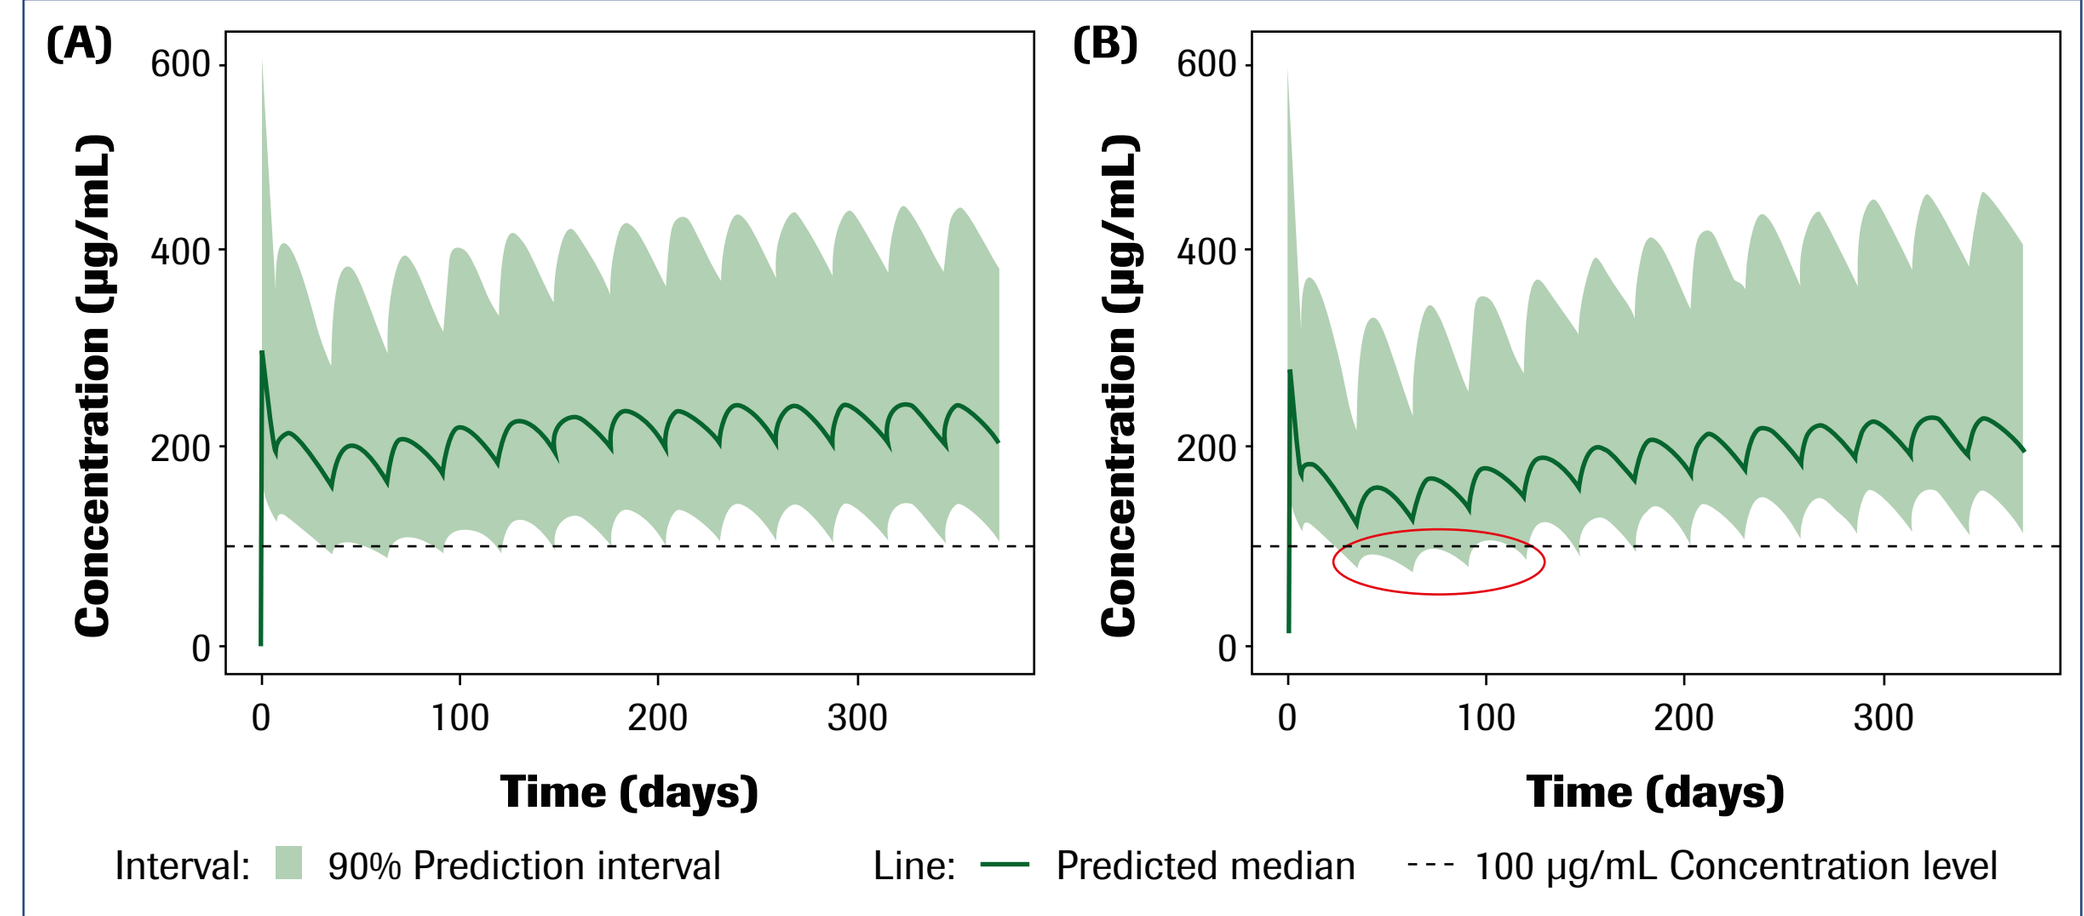
(A)
(B)
Concentration (μg/mL)
Time (days)
Interval: 90% Prediction interval
Line: Predicted median
100 μg/mL Concentration level

The red oval indicates where patients who switched from eculizumab were transiently at risk of having crovalimab fall below the target concentration.

Figure 9. Simulated concentration-time profiles of optimized crovalimab dose and regimen in (A) patients with PNH who were treatment naive; (B) patients with PNH who switched from eculizumab treatment

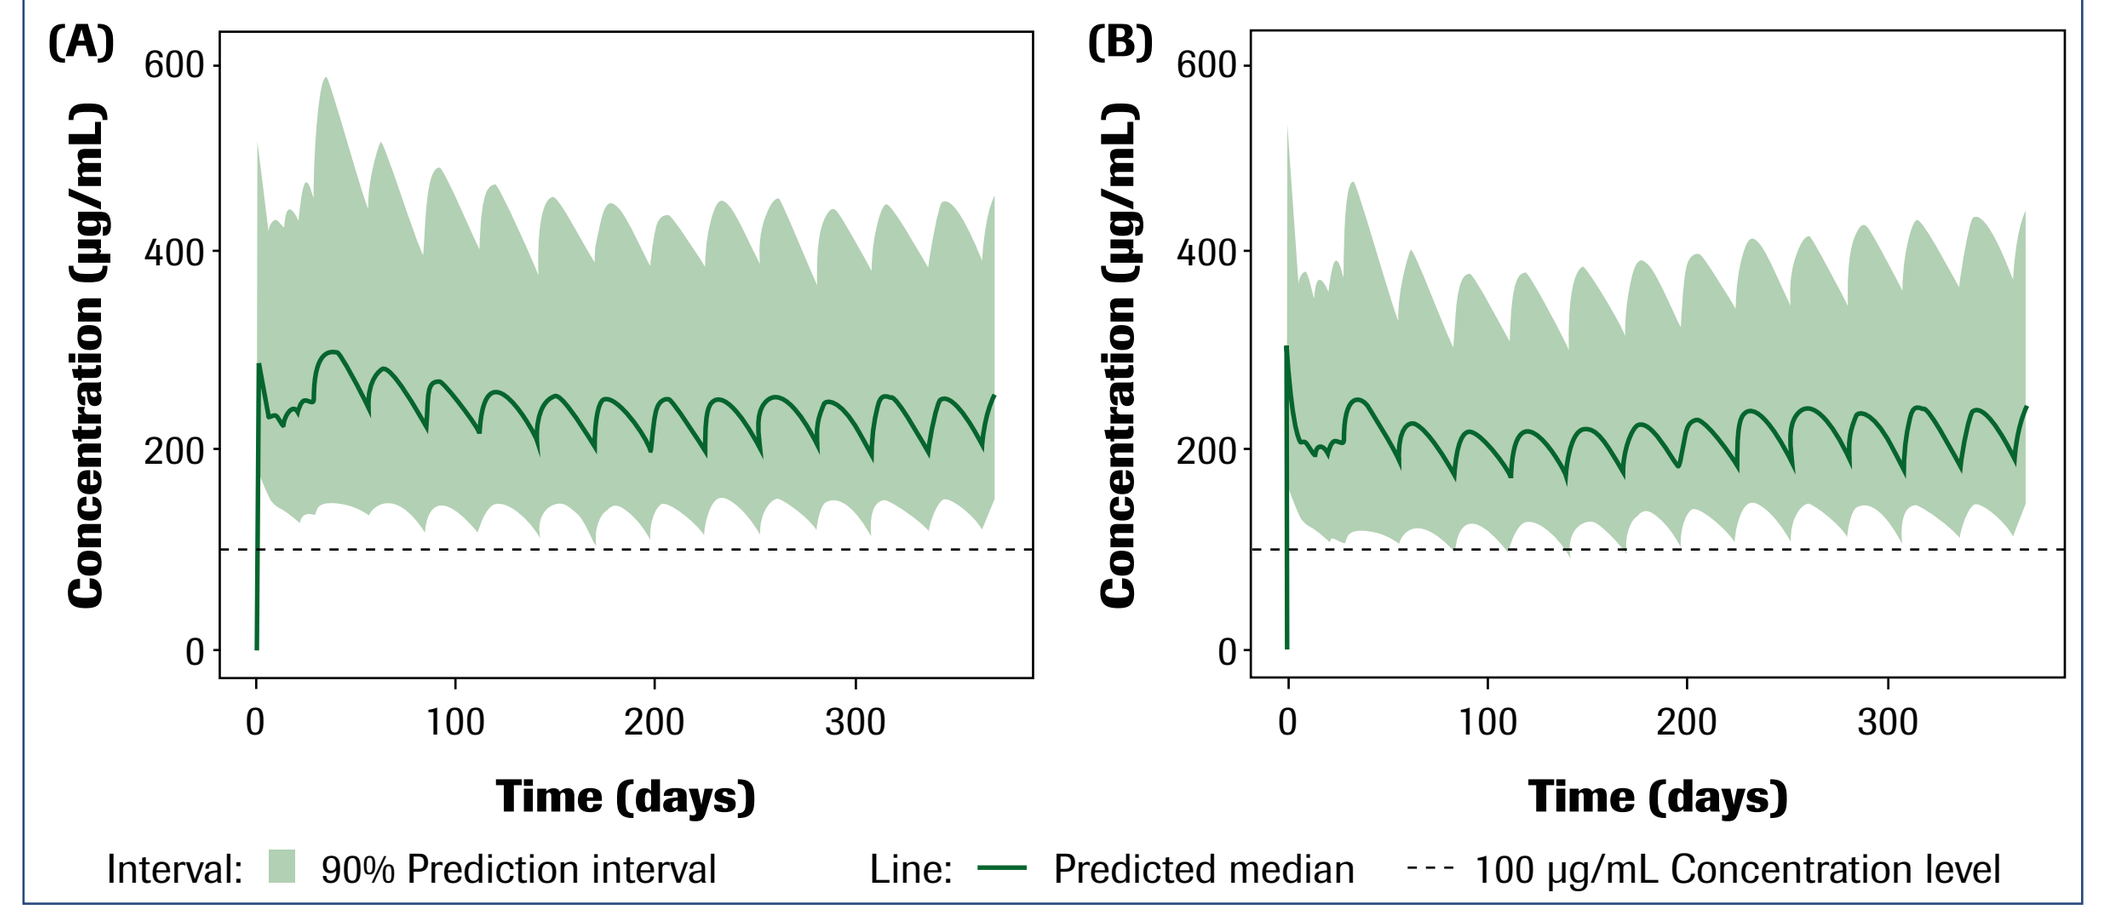
(A)
(B)
Concentration (μg/mL)
Time (days)
Interval: 90% Prediction interval
Line: Predicted median
100 μg/mL Concentration level

CONCLUSIONS

- PK/PD analyses showed that crovalimab had dose-proportional exposure, 100% bioavailability, and a terminal half-life of 30 days, enabling SC administration.
- Exposure-response characterization demonstrated the potential of crovalimab as an infrequently administered SC therapy for PNH.
- Approximately 100 μg/mL of crovalimab achieves complement inhibition.

REFERENCES

1. Brodsky RA. *Blood*. 2014;124(18):2804-2811; 2. Hillmen P, et al. *N Engl J Med*. 2006;355(12):1233-1243; 3. Kulasekararaj AG, et al. *Blood*. 2019;133(6):540-549; 4. Fukuzawa T, et al. *Sci Rep*. 2017;7(1):1080; 5. Nishimura J, et al. *N Engl J Med* 2014;370(7):632–639; 6. Sampei Z, et al. *PLoS One*. 2018;13(12):e0209509; 7. Sostelly A, et al. ASH 2019 [abstract 1227]; 8. Röth A, et al. *Blood*. 2017;130(suppl 1) [abstract 4750], 9. Röth A, et al. *Blood*. 2018; 132(suppl 1) [abstract 535].

ACKNOWLEDGMENTS

This study was sponsored by F. Hoffmann-La Roche Ltd, Basel, Switzerland. We thank the patients, their families, the nurses, the clinical study sites, and the investigators who participated in this study. Support for third-party writing assistance for this poster by Samantha Santangelo, PhD, of Health Interactions, Inc, was provided by F. Hoffmann-La Roche Ltd.

DISCLOSURES

Alexandre Sostelly, Simon Buatois, Christoph Bucher, and Jean Eric Charoin are employees of F. Hoffmann-La Roche. Antoine Soubret is an employee of F. Hoffmann-La Roche and owns stock in the company. Erica Winter has no conflicts to declare. Barbara Klughammer is an employee of F. Hoffmann-La Roche AG and owns stock in the company. Joy C Hsu is an employee of Roche/Genentech and owns stock in the company. Gregor Jordan is an employee of Roche Diagnostics GmbH and owns stock in the company. Keisuke Gotanda and Kenji Shinomiya are employees of Chugai Pharmaceutical Co, Ltd; Keisuke Gotanda also holds a patent for crovalimab. Zsolt Nagy sits on advisory boards for Janssen, Novartis, Takeda, AbbVie, and Roche. Jens Panse sits on advisory committees for Apellis, BMS, Grünenthal, MSD, and Roche; on speakers bureaus and advisory committees for Alexion, Boehringer Ingelheim, and Novartis; and on speakers bureaus for Pfizer and Chugai Pharmaceutical Co, Ltd. Sung-Soo Yoon is a consultant to Amgen, Novartis, and Janssen; received research funding from Kyowahako Kirin and Yuhan Pharma; and received honoraria from Novartis and Amgen. Régis Peffault de Latour is a consultant to Alexion, Pfizer, and Novartis; received research funding from Alexion, Pfizer, Novartis, and Amgen; and received honoraria from Alexion, Pfizer, and Novartis. Junichi Nishimura received research funding and honoraria from Alexion and is an advisor to Chugai Pharmaceutical Co, Ltd. Alexander Röth is a consultant to Alexion, Apellis, Novartis, Roche, and Bioverativ; received research funding from Roche; and received honoraria from Alexion.

PUSHED FOR TIME?

In order to use the QR code, please open the camera on your phone and hold it over the QR code or download an app called "QR code reader" from the Apple AppStore or the Android Google Play Store

Receive an instant copy of this poster

Request additional presentations of trials sponsored/supported by Roche at this congress

If you are having difficulties using the QR code reader, please visit https://medically.roche.com/